NUC-3373 potentiates immune-mediated cytotoxicity of CRC Cells

Oliver J. Read[1,2], Jennifer Bré[1,2], David J. Harrison[1,2]
1) University of St Andrews, St Andrews UK
2) NuCana plc, Edinburgh, UK

Abstract Number: **1113**

Email: **oliverread@nucana.com**

BACKGROUND

NUC-3373 is a potent TS inhibitor with a DNA-mediated mode of action

- ProTide transformation of FUDR-MP[1,2], the active anti-cancer metabolite of 5-FU
- Designed to overcome the key 5-FU resistance mechanisms
 - Protected from breakdown by DPD
 - FUDR-MP generation independent of intracellular enzymatic activation
- NUC-3373 generates significantly higher levels of FUDR-MP compared to 5-FU[3,4]
- NUC-3373 is currently being investigated in patients with advanced CRC (NuTide:302 Phase Ib/II; NCT03428958)
- Interaction between cancer cells and immune cells in the microenvironment is important for anticancer activity

NUC-3373 induces an anti-tumor inflammatory response by:

- Inducing ER stress via accumulation of TS ternary complexes[5]
- Releasing DAMPs (CRT, HSP70, ATP & HMGB1)
- Activating the innate immune system (NK cell response)[6]

Aim:

To confirm that NUC-3373 enhances immunogenic cell death (ICD) via the adaptive immune system and that it can potentiate the effects of PD-1/PD-L1 (e.g. nivolumab)

Hypotheses:

- NUC-3373-induced DAMPs can also promote activation of the adaptive immune system leading to ICD
- NUC-3373 potentiates the effects of PD-1 inhibitor nivolumab

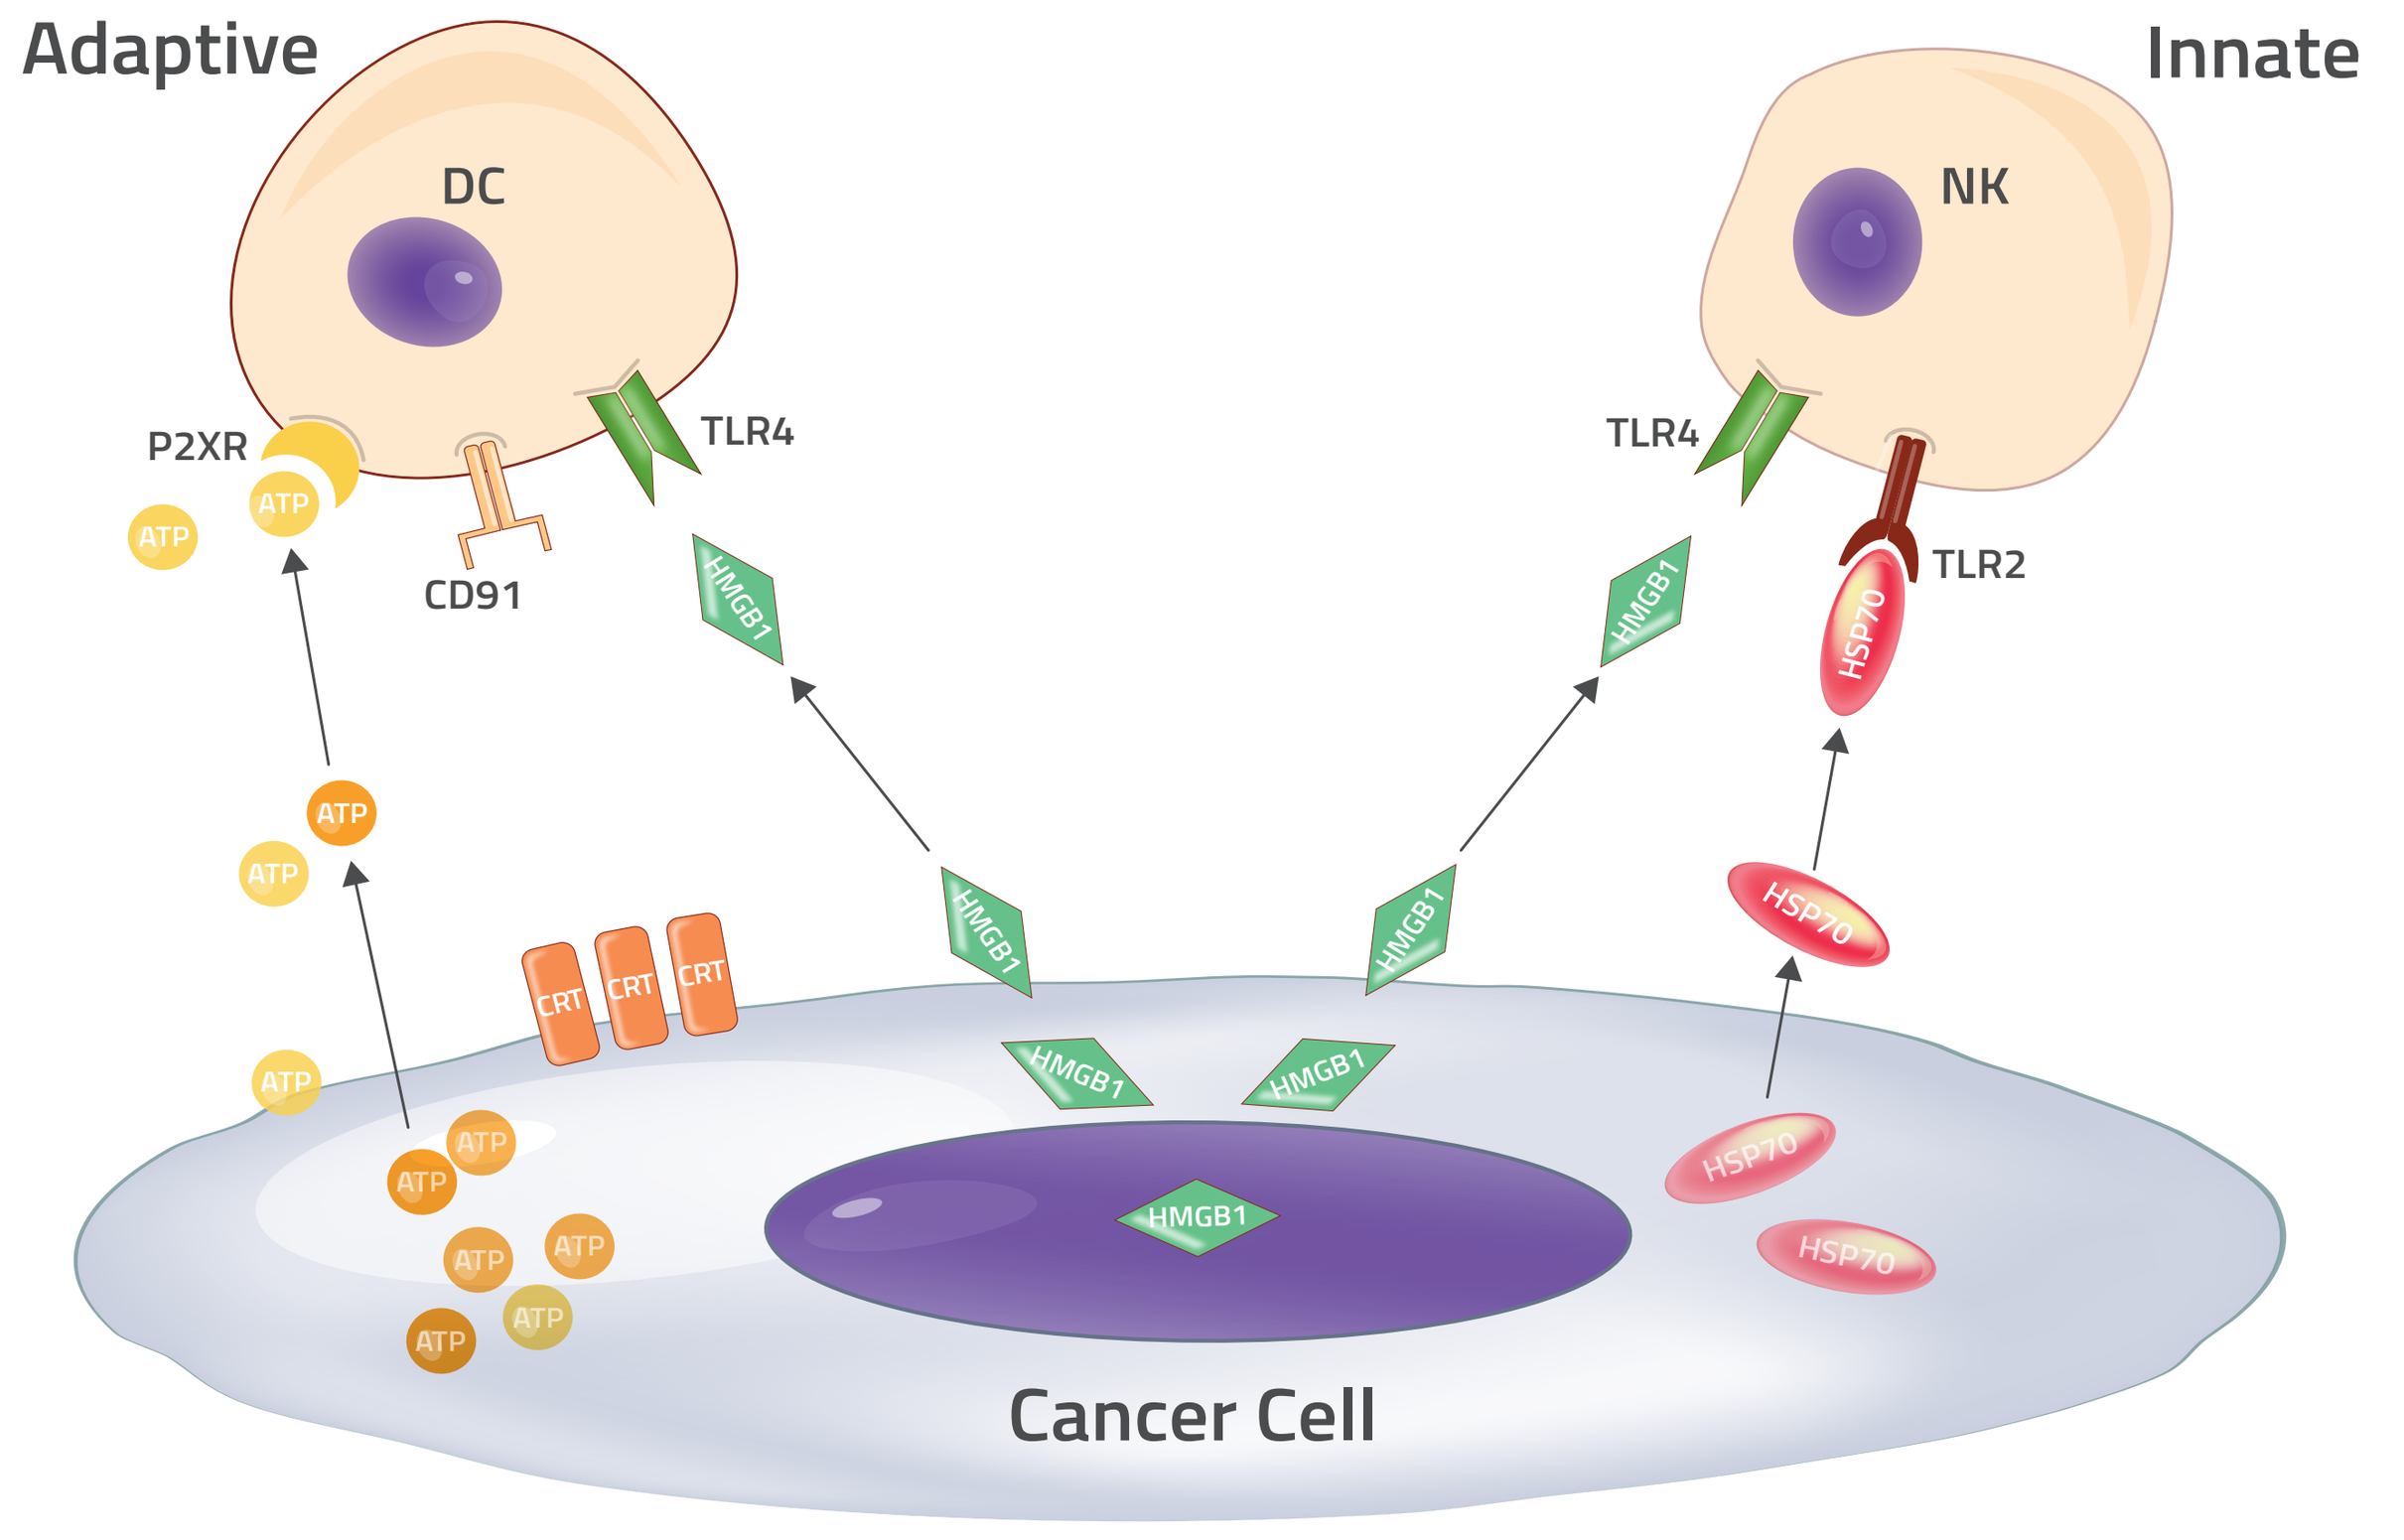
Adaptive
Innate
DC
NK
P2XR
ATP
CD91
TLR4
TLR2
HMGB1
HSP70
CRT
Cancer Cell

Figure 1. DAMPs release from CRC cells treated with NUC-3373

METHODS

Cell culture

CRC cell lines HCT116 (MSI) and SW480 (MSS) were treated with sub-IC_{50} doses of NUC-3373 for 24 hours prior to coculture with PBMCs (IC_{50}: HCT116 = 25 µM, SW480 = 50 µM)
An anti-CD3/28 monoclonal antibody (mAb)-stimulated PBMCs model was used to test T-cell mediated cell killing modulated by NUC-3373 ± nivolumab (10 µg/ml)

Pro-inflammatory cytokine profile

qPCR was used to assess the gene expression of pro-inflammatory cytokines

PD-L1 expression

PD-L1 surface expression on CRC cells and PBMCs was determined by flow cytometry

Cytotoxicity

Cell viability was measured by automated cytometry analysis (Celigo) and sulforhodamine B assay

RESULTS

NUC-3373 induces pro-inflammatory cytokines

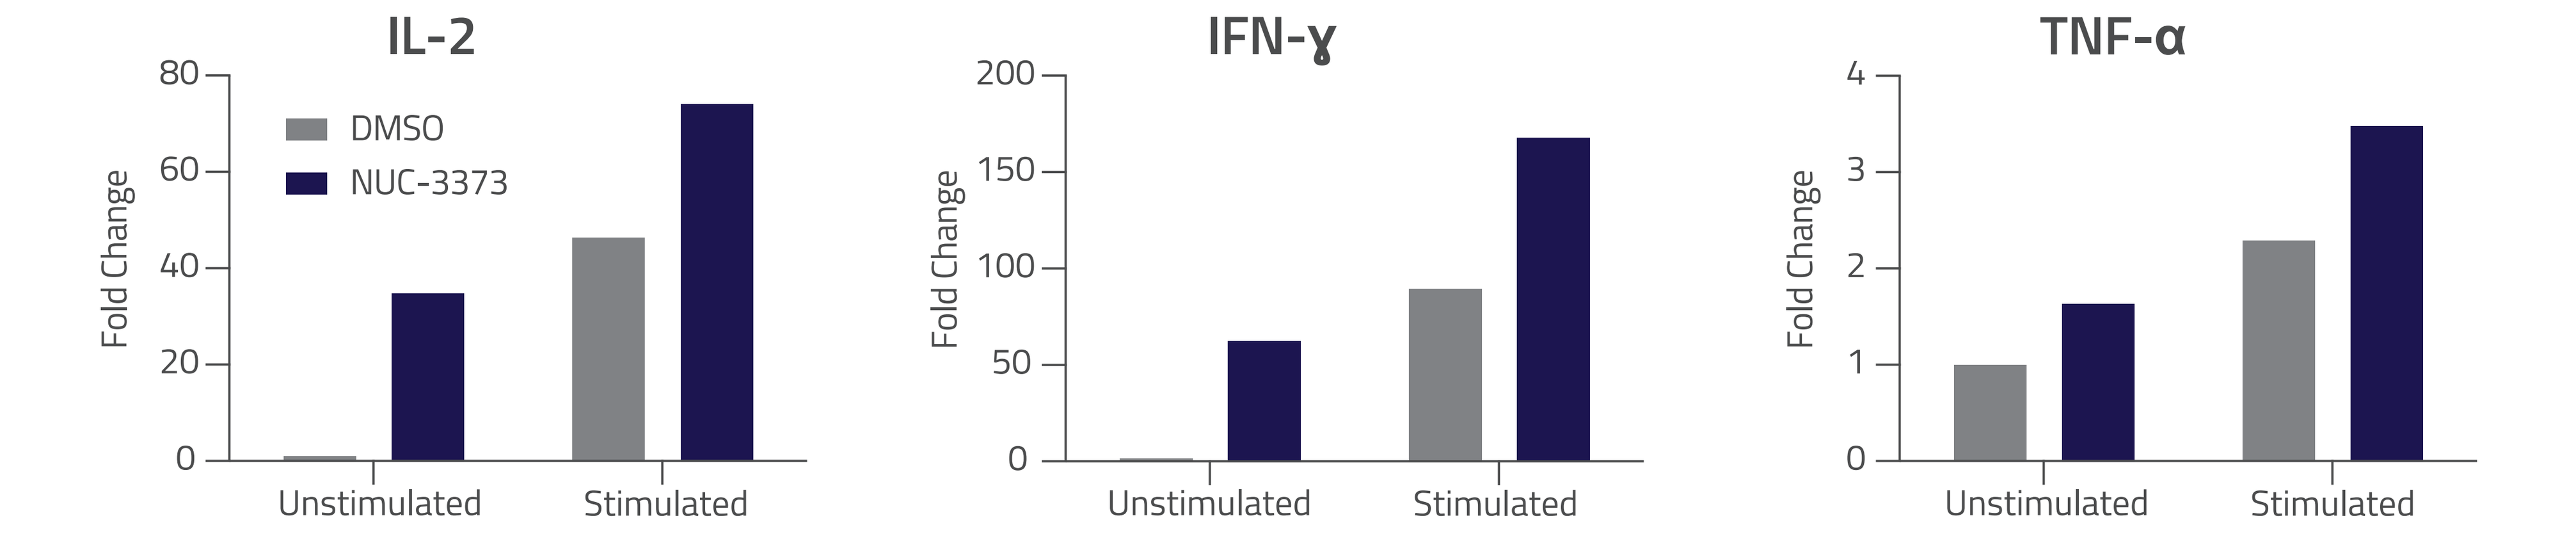
IL-2
IFN-ɣ
TNF-α
DMSO
NUC-3373
Fold Change
Unstimulated
Stimulated

Figure 2. Representative graphs showing effect of 10 µM NUC-3373 (± PBMC stimulation in coculture) on gene expression over 24 hours

- NUC-3373 increases gene expression of pro-inflammatory cytokines
 - Effect potentiated by PBMC stimulation

NUC-3373 induces PD-L1 expression on CRC and immune cells

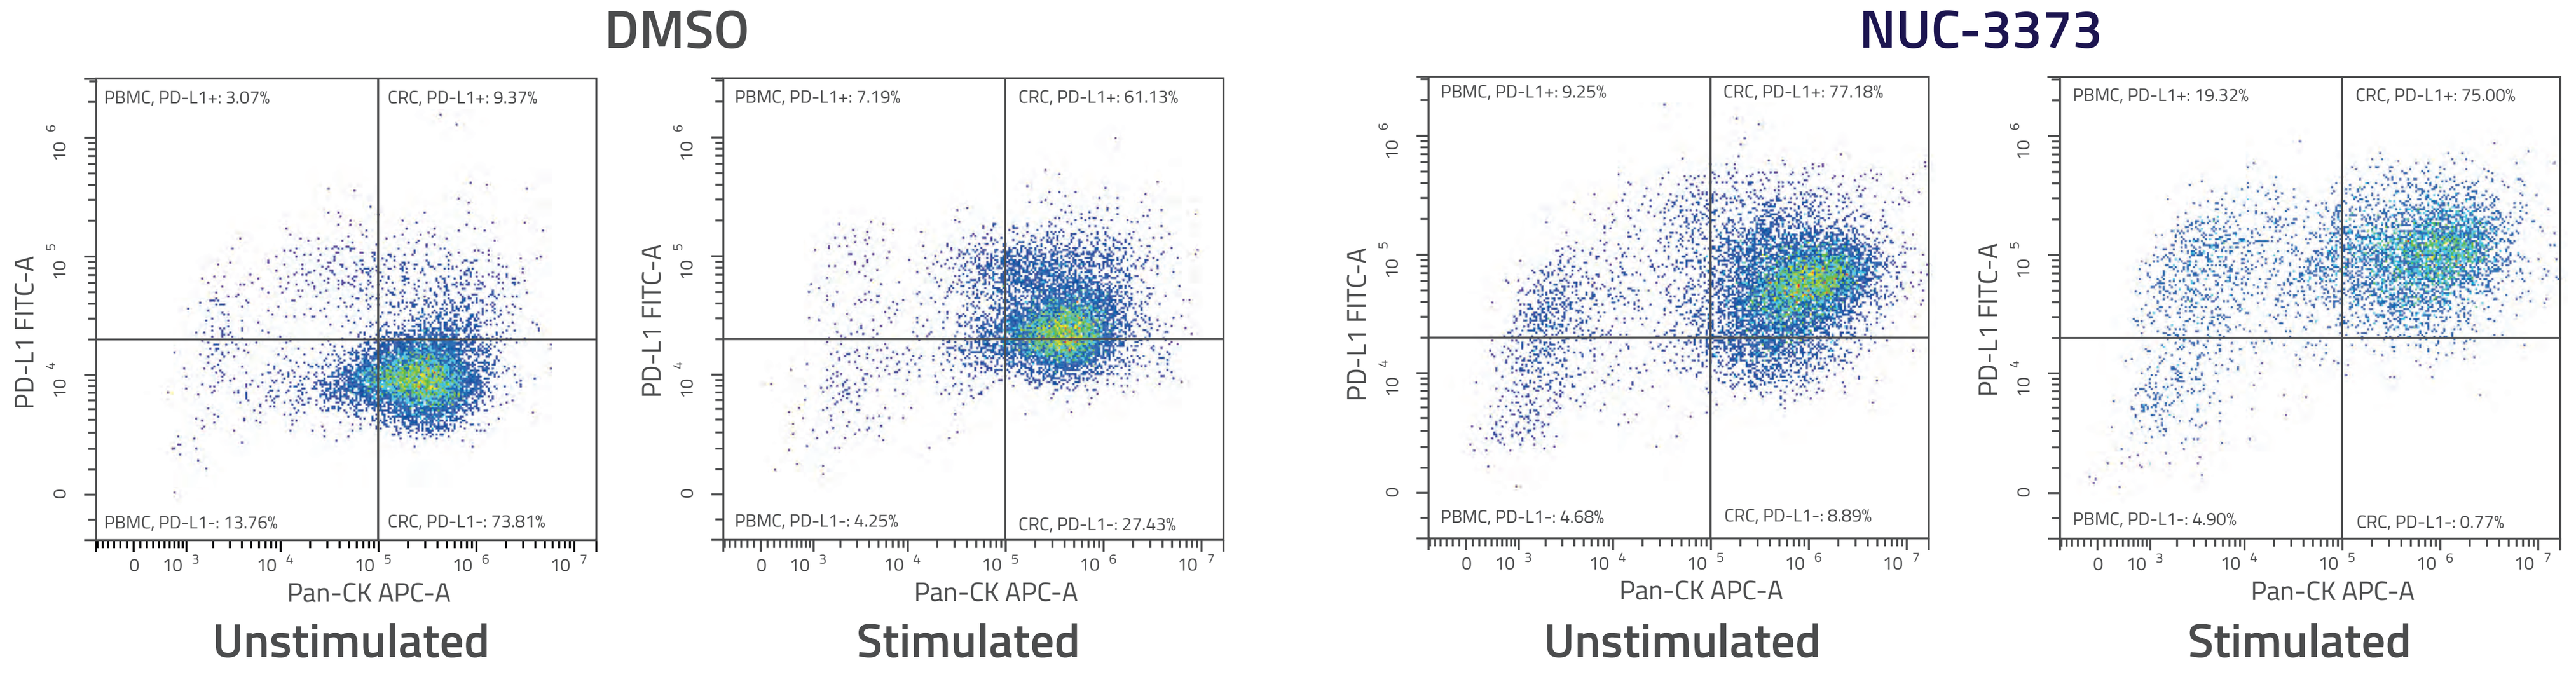
DMSO
NUC-3373
PBMC, PD-L1+: 3.07%
CRC, PD-L1+: 9.37%
PBMC, PD-L1-: 13.76%
CRC, PD-L1-: 73.81%
PBMC, PD-L1+: 7.19%
CRC, PD-L1+: 61.13%
PBMC, PD-L1-: 4.25%
CRC, PD-L1-: 27.43%
PBMC, PD-L1+: 9.25%
CRC, PD-L1+: 77.18%
PBMC, PD-L1-: 4.68%
CRC, PD-L1-: 8.89%
PBMC, PD-L1+: 19.32%
CRC, PD-L1+: 75.00%
PBMC, PD-L1-: 4.90%
CRC, PD-L1-: 0.77%
PD-L1 FITC-A
Pan-CK APC-A
Unstimulated
Stimulated
Unstimulated
Stimulated

Figure 3a. PD-L1 surface expression on HCT116 CRC cells pre-treated with 10 µM NUC-3373 and cocultured with PBMCs after 48 hours

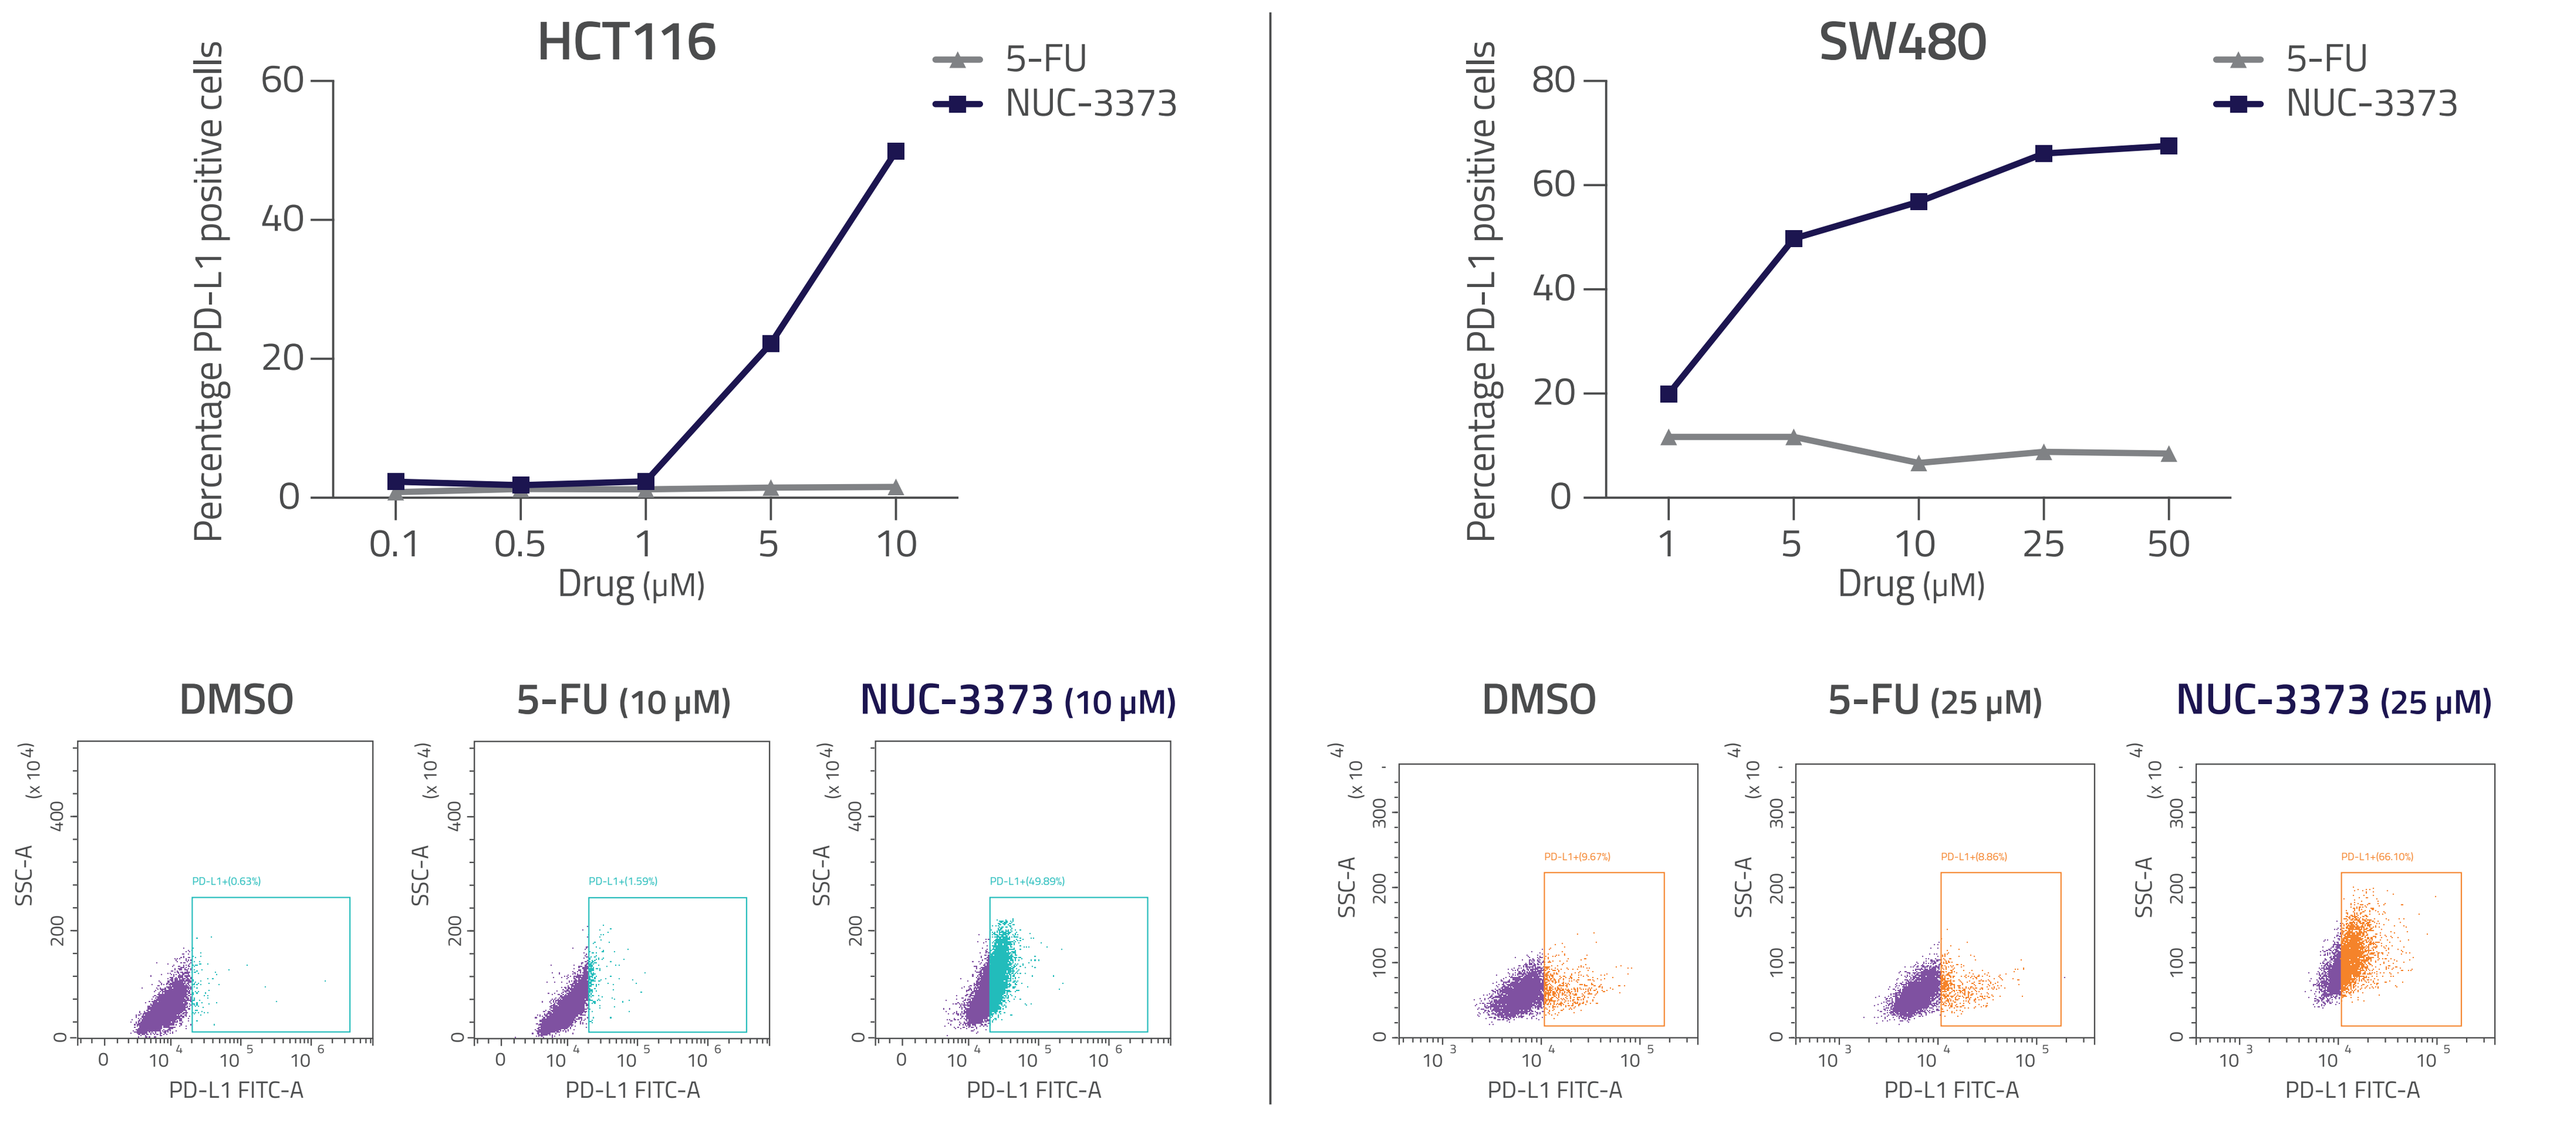
HCT116
SW480
5-FU
NUC-3373
Percentage PD-L1 positive cells
Drug (µM)
DMSO
5-FU (10 µM)
NUC-3373 (10 µM)
DMSO
5-FU (25 µM)
NUC-3373 (25 µM)
SSC-A
PD-L1 FITC-A

Figure 3b. PD-L1 surface expression in HCT116 (MSI) and SW480 (MSS) CRC cells treated with 5-FU or NUC-3373

- NUC-3373 increased PD-L1 surface expression on CRC cells and PBMCs
 - Effect observed regardless of PBMC stimulation
- NUC-3373 increased PD-L1 surface expression on CRC cells in the absence of PBMCs
 - Independent of MSI status
- 5-FU had no effect on PD-L1 surface expression (equimolar doses)
- These data suggest that PD-L1 induction is NUC-3373 mediated

NUC-3373 enhances immunogenic cell death

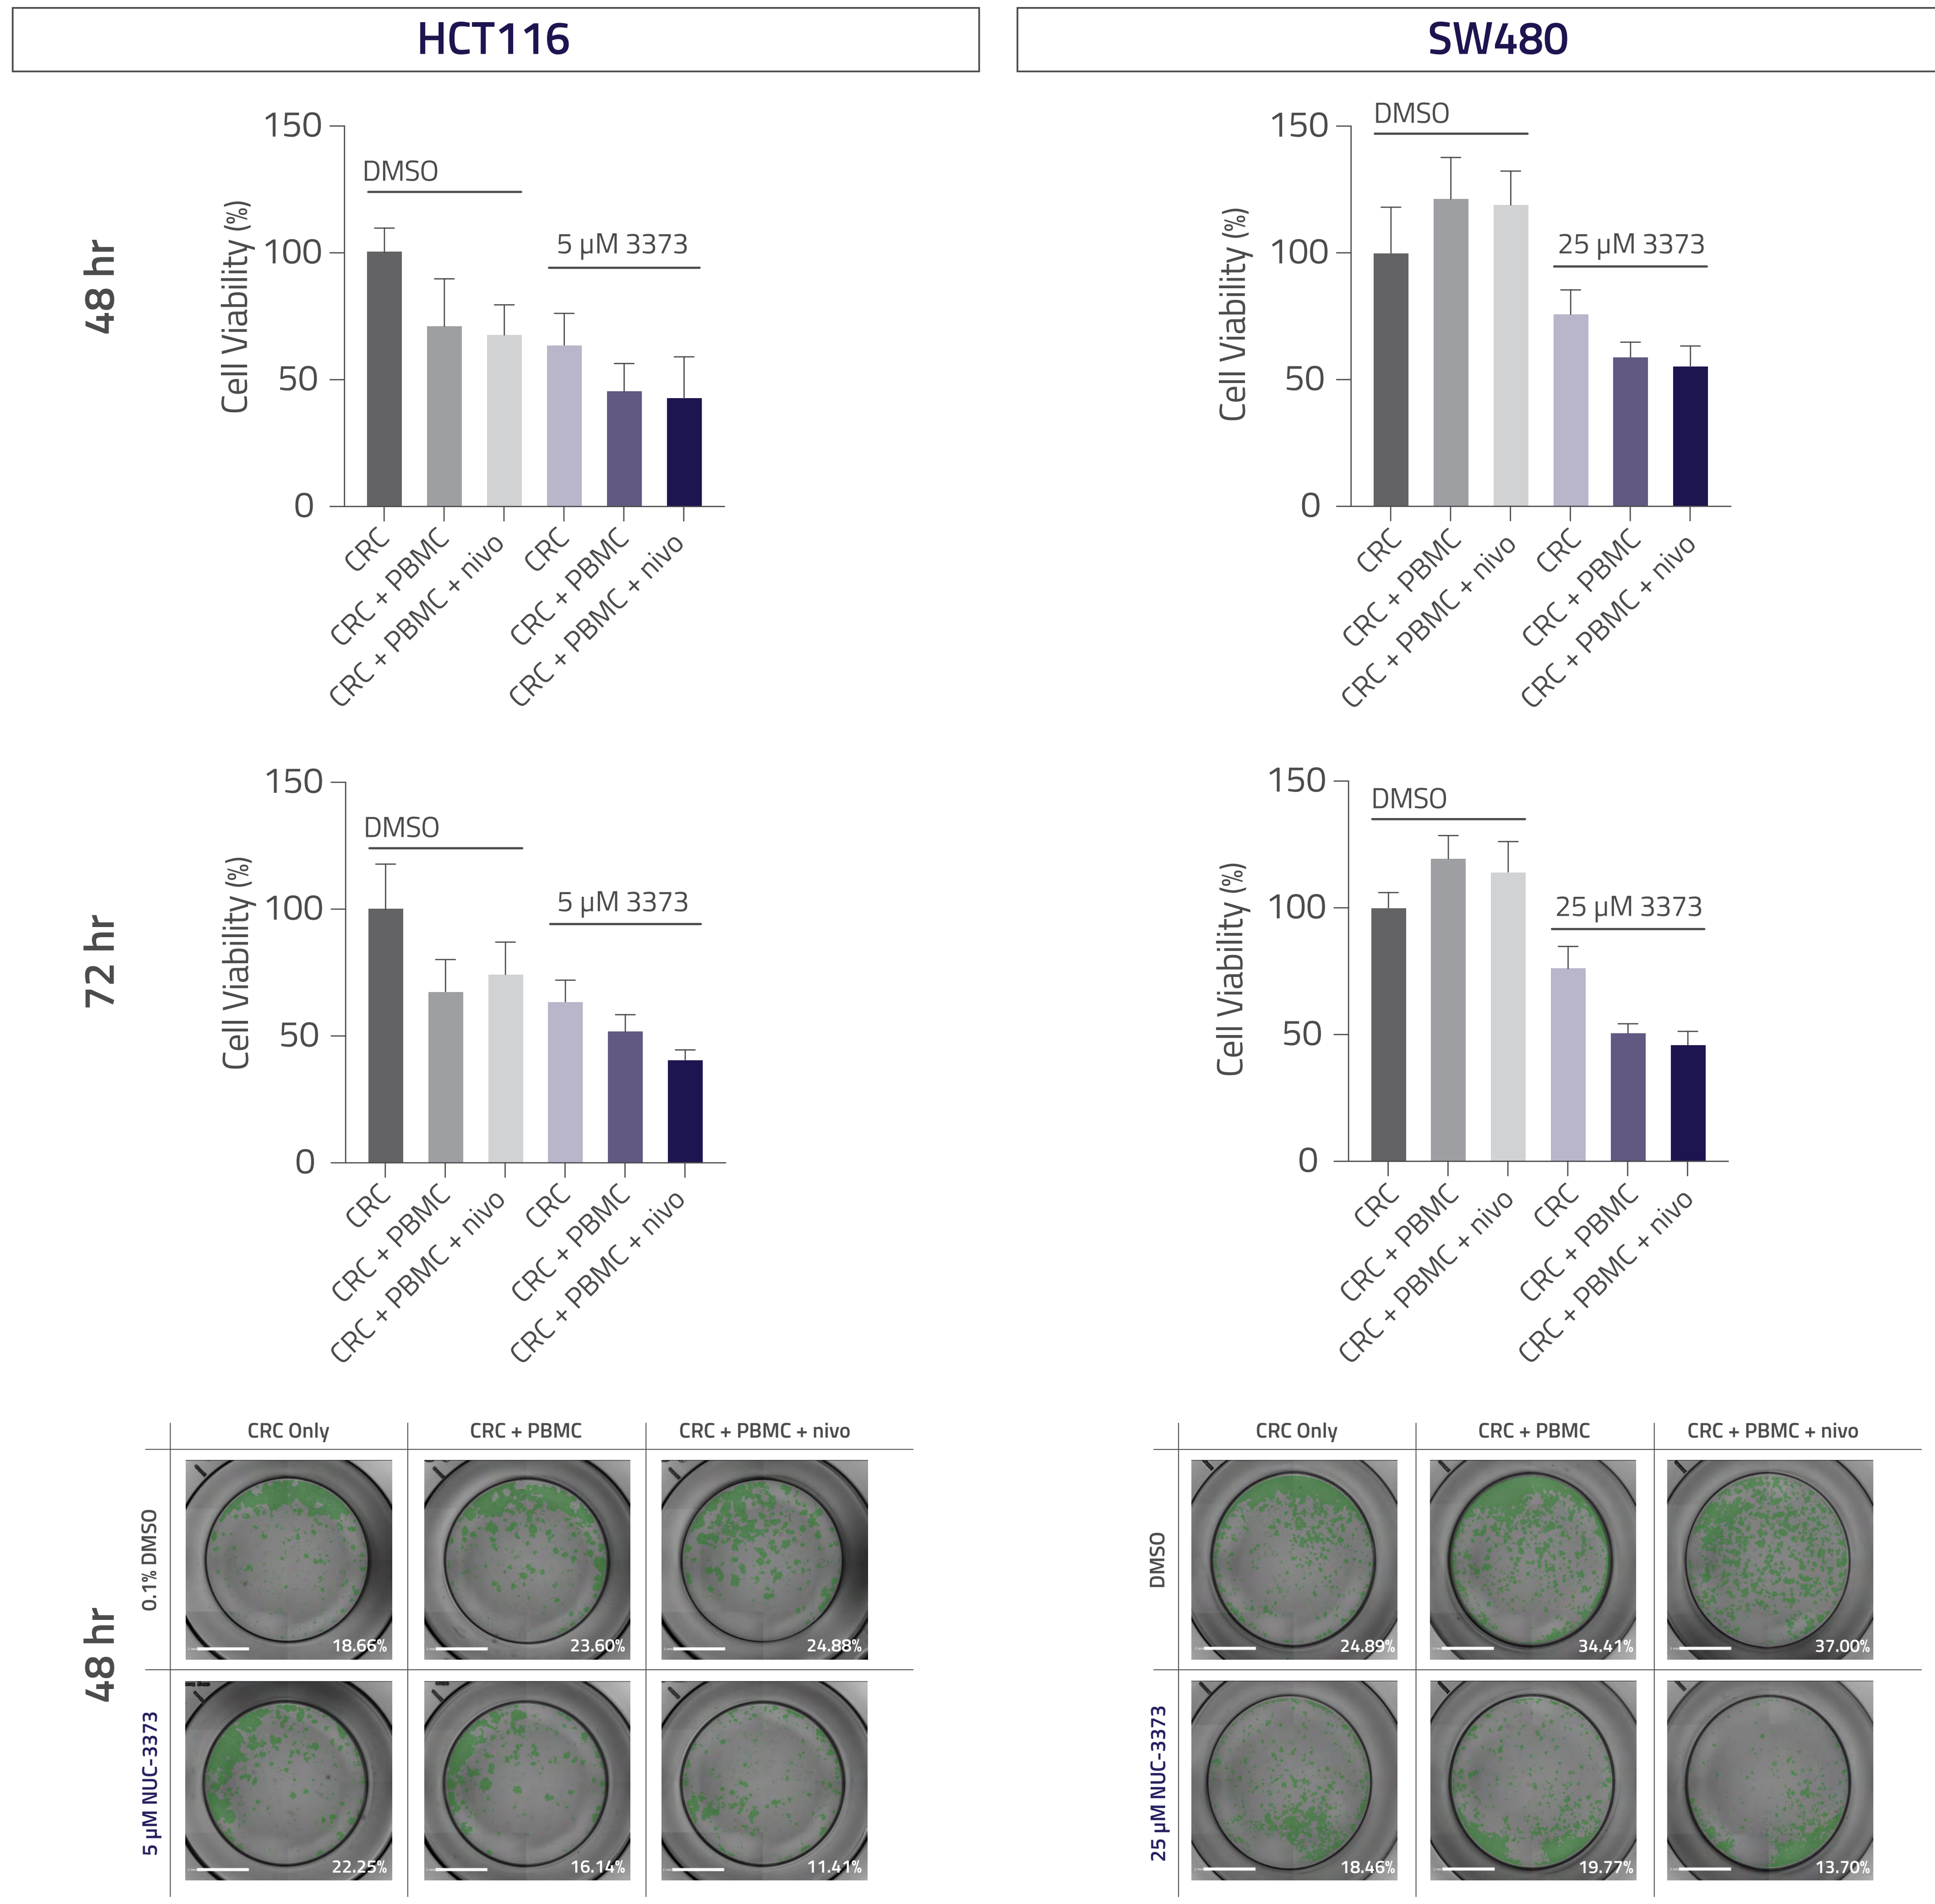
HCT116
SW480
48 hr
72 hr
Cell Viability (%)
DMSO
5 µM 3373
25 µM 3373
CRC
CRC + PBMC
CRC + PBMC + nivo
CRC Only
CRC + PBMC
CRC + PBMC + nivo
0.1% DMSO
5 µM NUC-3373
DMSO
25 µM NUC-3373
18.66%
23.60%
24.88%
22.25%
16.14%
11.41%
24.89%
34.41%
37.00%
18.46%
19.77%
13.70%

Figure 4. Cell viability of HCT116 (MSI) and SW480 (MSS) CRC cells pre-treated with NUC-3373 and cultured with PBMCs

- NUC-3373 pre-treatment of CRC cells:
 - Enhances ICD caused by PBMCs regardless of MSI status
 - Potentiates the effect of nivolumab

CONCLUSION

- **NUC-3373 induces DAMPs & pro-inflammatory cytokines**
 - **Promoting an anti-cancer immunological environment**
- **NUC-3373 induces PD-L1 expression on CRC & immune cells**
- **NUC-3373 evokes immunogenic cell death & potentiates the effect of nivolumab**

NUC-3373 is an attractive combination partner for PD-1/PD-L1 inhibitors

REFERENCES: 1. Vande Voorde J *et al.*, 2011 *Biochem Pharmacol*; 82: 441-452 2. McGuigan C *et al.*, 2011 *Med Chem*; 27: 7247-7258 3. Ghazaly E *et al.*, 2017 Ann Oncol; Suppl_5128 4. Derissen EJ *et al.*, 2016 *Clin Pharmacol*; 81: 949-957 5. McKissock FG *et al.*, 2020 *Cancer Res*; 80 (Suppl_16); 1848 6. Read OJ *et al.*, 2021 *Cancer Res*; 81 (Suppl_13); 1655

ABBREVIATIONS: 5-FU: 5-fluorouracil, APC-A: Allophycocyanin, ATP: Adenosine triphosphate, CD91: α2-macroglobulin receptor, CRT: Calreticulin, CRC: Colorectal cancer, DAMPs: Damage associated molecular patterns, DPD: Dihydropyrimidine dehydrogenase, DMSO: Dimethyl sulfoxide, DNA:Deoxyribonucleic Acid, DC: Dendritic cell, ER: Endoplasmic reticulum, FITC-A: Fluorescein isothiocyanate, FUDR-MP: Fluorodeoxyuridine monophosphate, HMGB1: High mobility group box 1, HSP-70: Heat shock protein 70, ICD: Immunogenic cell death, IFN-ɣ: Interferon gamma, IL-2: Interleukin 2, MSI:Microsatellite instability, MSS:Microsatellite stabile, NK: Natural killer, P2XR: P2X receptor, Pan-CK: Pan cytokeratin, PBMC: Peripheral blood mononuclear cells, PD-1: Programmed cell death receptor 1, PD-L1: Programmed death ligand 1, qPCR: Quantitative polymerase chain reaction, SSC-A: Side scatter, TLR:Toll-like receptor, TNF-α: Tumor necrosis factor alpha, TS: Thymidylate synthase